Nuclear regulatory landscape of the circadian clock in mouse liver

Jingkui Wang[1], Daniel Mauvoisin[2], Eva Martin[2], Florian Atger[2], Antonio Nunes Galindo[2], Federico Sizzano[2], Loïc Dayon[2], Martin Kussmann[2], Patrice Waridel[3], Manfredo Quadroni[3], Felix Naef[1] and Frédéric Gachon[2]

[1]Institute of Bioengineering, School of Life Sciences, Ecole Polytechnique Fédérale de Lausanne and Swiss Institute of Bioinformatics, Lausanne, Switzerland
[2]Nestlé Institute of Health Sciences, Lausanne, Switzerland.
[3]Protein Analysis Facility, University of Lausanne, Lausanne, Switzerland.

Abstract

Diurnal oscillations of gene expression dictated by the circadian clock enable living organisms to coordinate their physiological processes with daily environmental changes. Although such rhythms have been extensively studied at the level of transcription and mRNA accumulation, comparably little is known at the proteins level, though recent proteomics studies indicated that total protein rhythms generally appeared damped compared to their cognate mRNAs. In order to further dissect how diurnal rhythms affect key functions such as transcription or chromatin remodeling, we quantified the temporal nuclear accumulation of proteins and phosphoproteins from mouse liver by SILAC-based MS. Our analysis identified ~5000 nuclear proteins, including all core-clock and clock-related proteins, over 500 of which are found to be rhythmic under a stringent statistical threshold (FDR <5%). These rhythmic nuclear proteins are mainly controlled at the post-transcriptional level and are often parts of complexes showing robust diurnal nuclear accumulation. These rhythmic complexes are notably involved in transcriptional regulation, rRNA synthesis, ribosome assembly, as well as DNA damage repair. From the parallel analysis of the nuclear phospho-proteome, we could infer the temporal activity of kinases contributing to these rhythmic phosphorylations. A large fraction of the kinase activities were implicated in cell signaling and cell cycle regulation. In addition, 80 transcription factors and about 100 transcriptional coregulators showed clear diurnal oscillations in the nucleus, enlarging the extent of transcriptional and epigenetic regulations by the circadian clock and/or systemic cues. Finally, a number of proteins with functions in the cytoplasm are detected in the nucleus at a common and sharp time near the night-day transition. This phenomenon is probably linked to the rhythmic endoreplication occurring in hepatic cells and associated to leakage of the nuclear membrane. Taken together, these findings provide unprecedented insights into the regulatory landscape of the diurnal liver nucleus.

Previous work

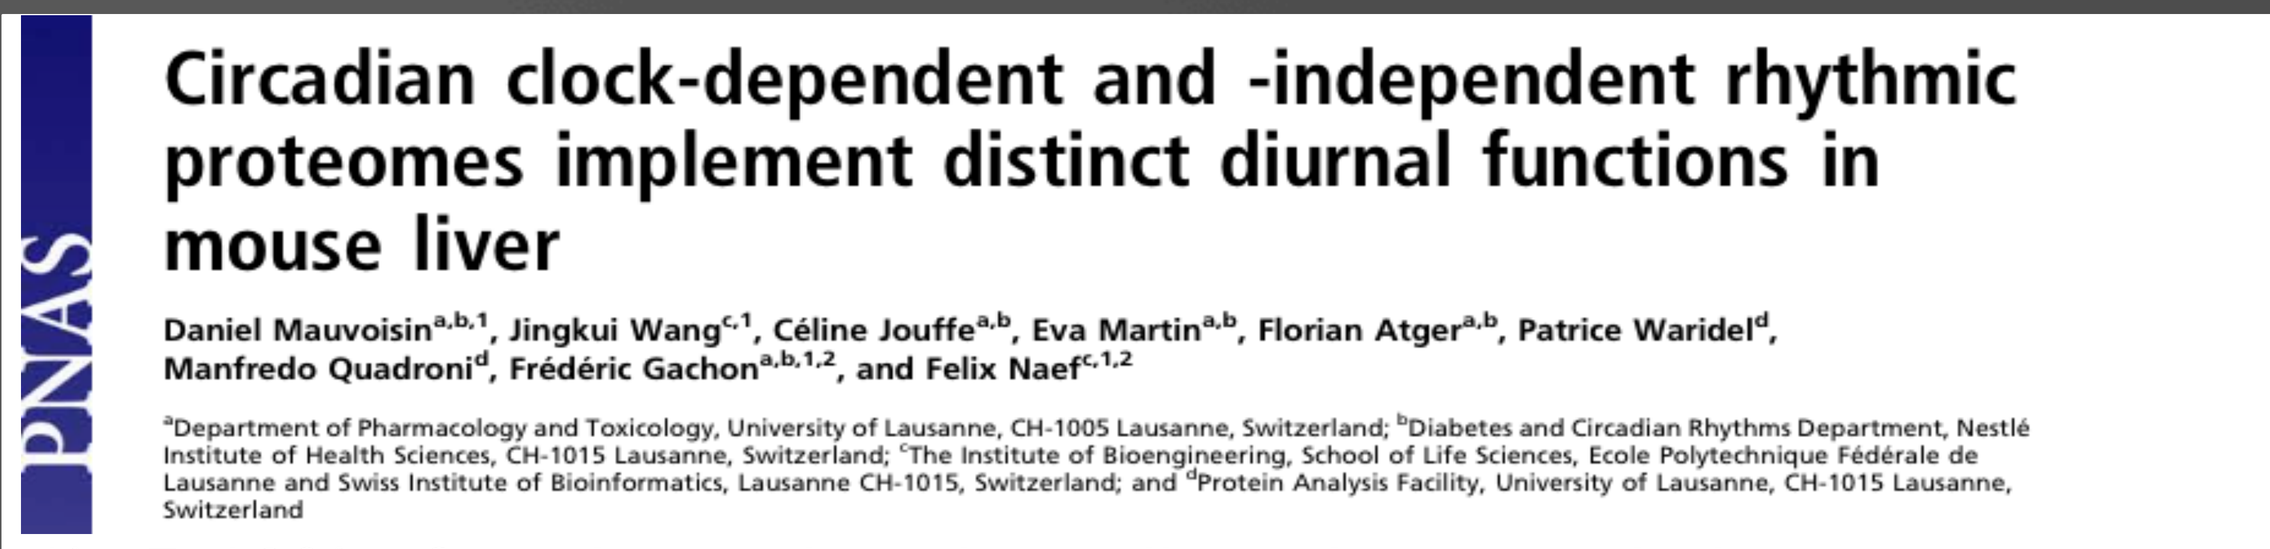
Circadian clock-dependent and -independent rhythmic proteomes implement distinct diurnal functions in mouse liver

Daniel Mauvoisin[a,b,1], Jingkui Wang[c,1], Céline Jouffe[a,b], Eva Martin[a,b], Florian Atger[a,b], Patrice Waridel[d], Manfredo Quadroni[d], Frédéric Gachon[a,b,1,2], and Felix Naef[c,1,2]

[a]Department of Pharmacology and Toxicology, University of Lausanne, CH-1005 Lausanne, Switzerland; [b]Diabetes and Circadian Rhythms Department, Nestlé Institute of Health Sciences, CH-1015 Lausanne, Switzerland; [c]The Institute of Bioengineering, School of Life Sciences, Ecole Polytechnique Fédérale de Lausanne and Swiss Institute of Bioinformatics, Lausanne CH-1015, Switzerland; and [d]Protein Analysis Facility, University of Lausanne, CH-1015 Lausanne, Switzerland

1) 5-10% of total proteins show diurnal accumulations.
2) ~50% of these rhythmic proteins do not have corresponding rhythmic mRNAs and are highly enriched in secretory proteins.

<u>**Question: How about the rhythmicity of proteins in different subcompartments such as nucleus ?**</u>

Methods & Results

SILAC mass spectrometry analysis of mouse liver nuclei

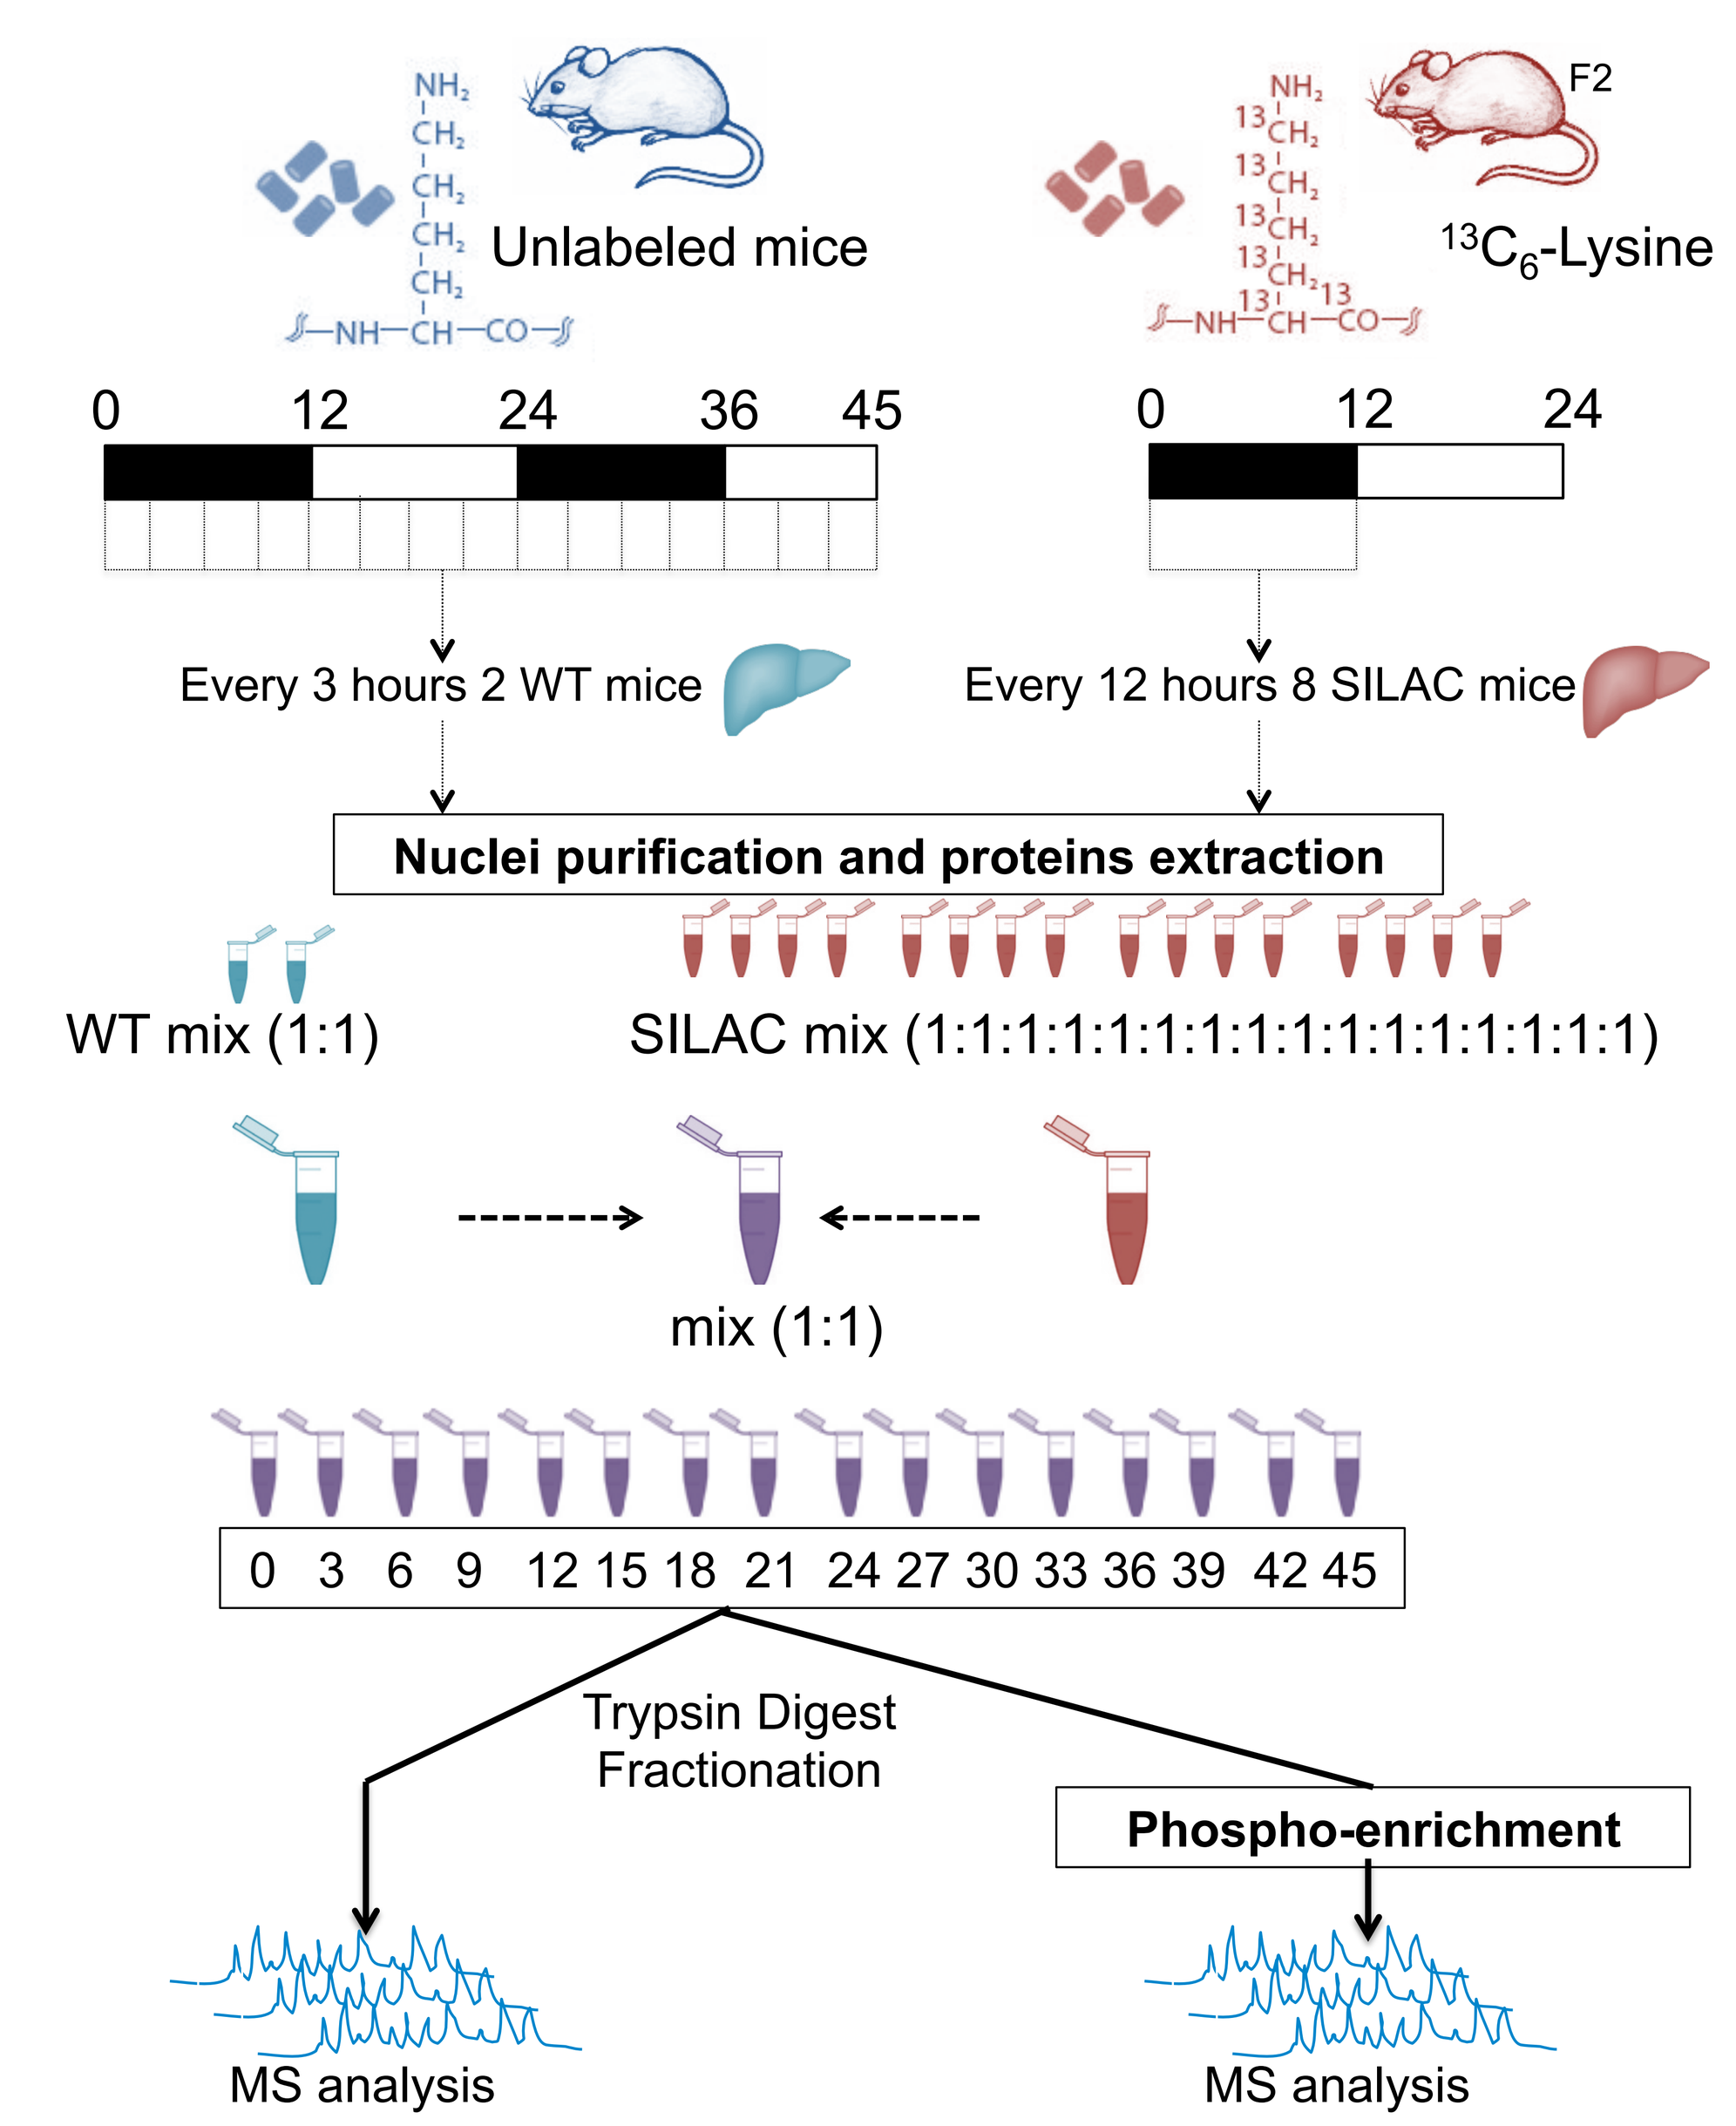
Unlabeled mice
F2
$^{13}C_6$-Lysine
Every 3 hours 2 WT mice
Every 12 hours 8 SILAC mice
Nuclei purification and proteins extraction
WT mix (1:1)
SILAC mix (1:1:1:1:1:1:1:1:1:1:1:1:1:1:1:1)
mix (1:1)
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45
Trypsin Digest
Fractionation
Phospho-enrichment
MS analysis
MS analysis

Fig. 1 ~ 5000 nuclear proteins were quantified by SILAC-based MS including core-clock and clock-related proteins

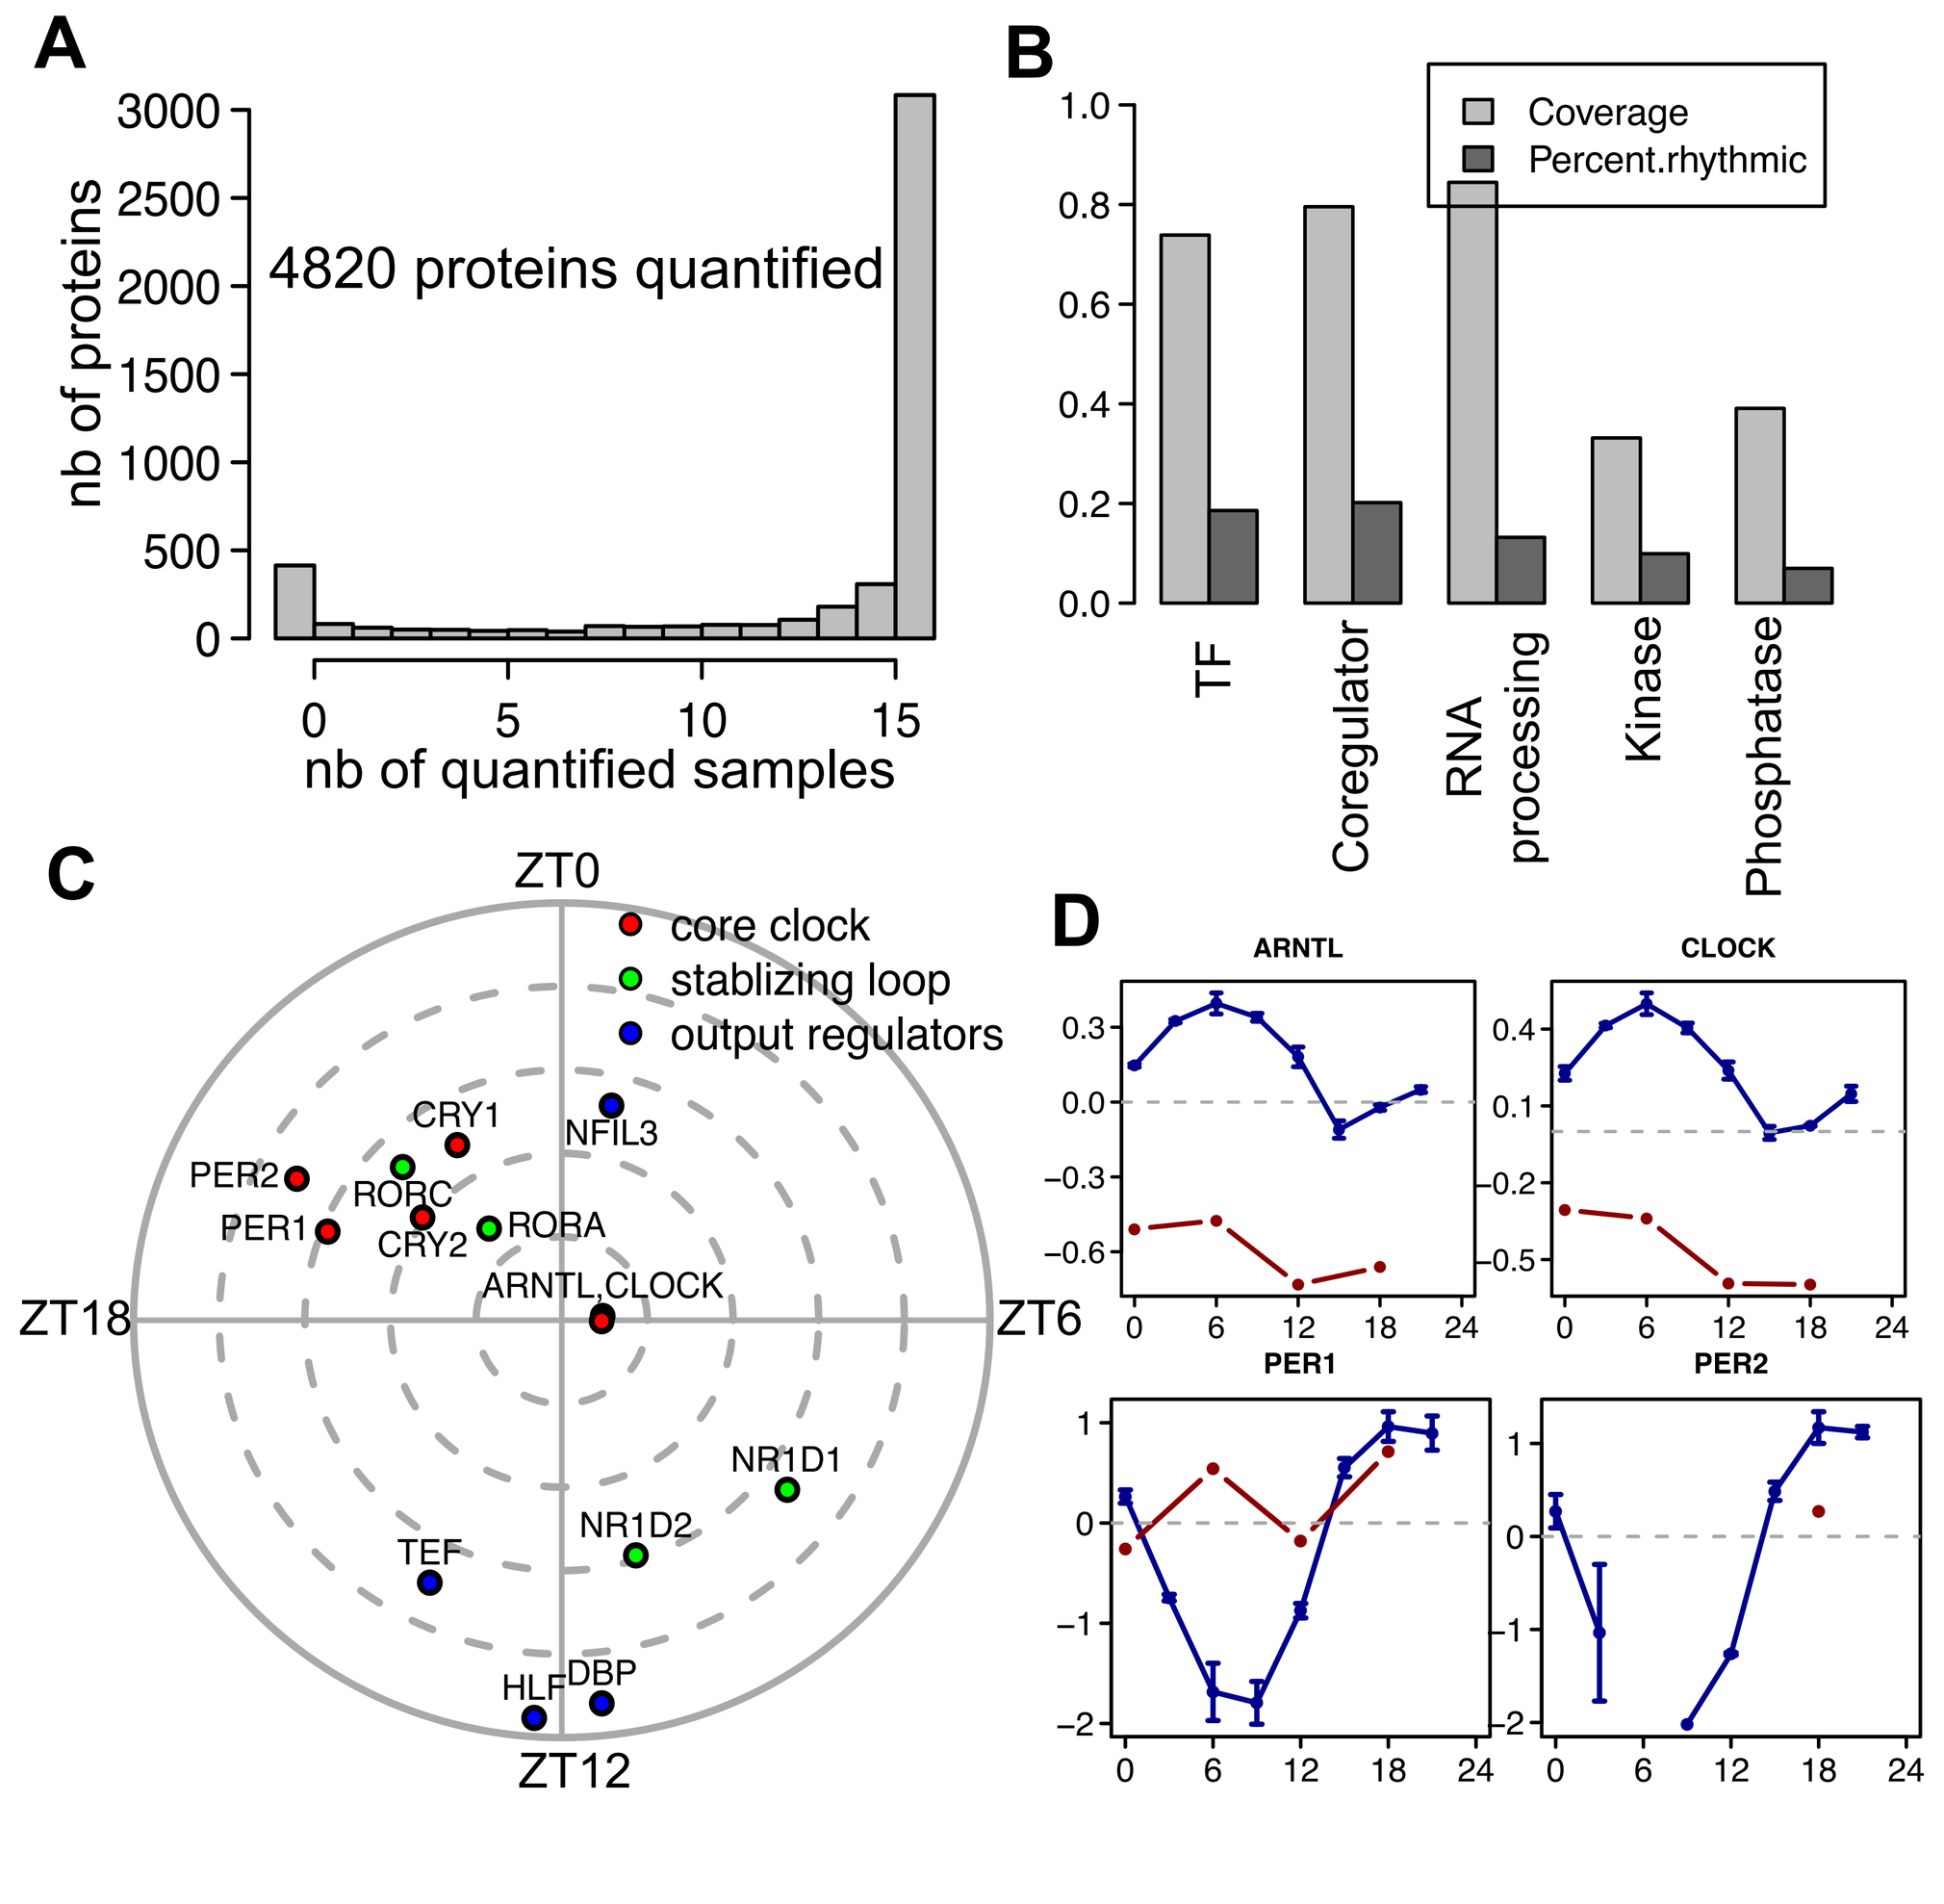
4820 proteins quantified
nb of proteins
nb of quantified samples
Coverage
Percent.rhythmic
TF
Coregulator
RNA processing
Kinase
Phosphatase
core clock
stablizing loop
output regulators
ZT0
ZT6
ZT12
ZT18
ARNTL
CLOCK
PER1
PER2

A) Distribution of samples numbers quantified for detected nuclear proteins.
B) Coverage of nuclear proteins and percentages of rhythmic ones (FDR<5%) for different functional categories.
C) Phases and peak-trough amplitudes of core-clock and clock-related proteins with examples in **D).**

Fig. 2 Rhythmic nuclear proteins are mainly post-transcriptionally regulated

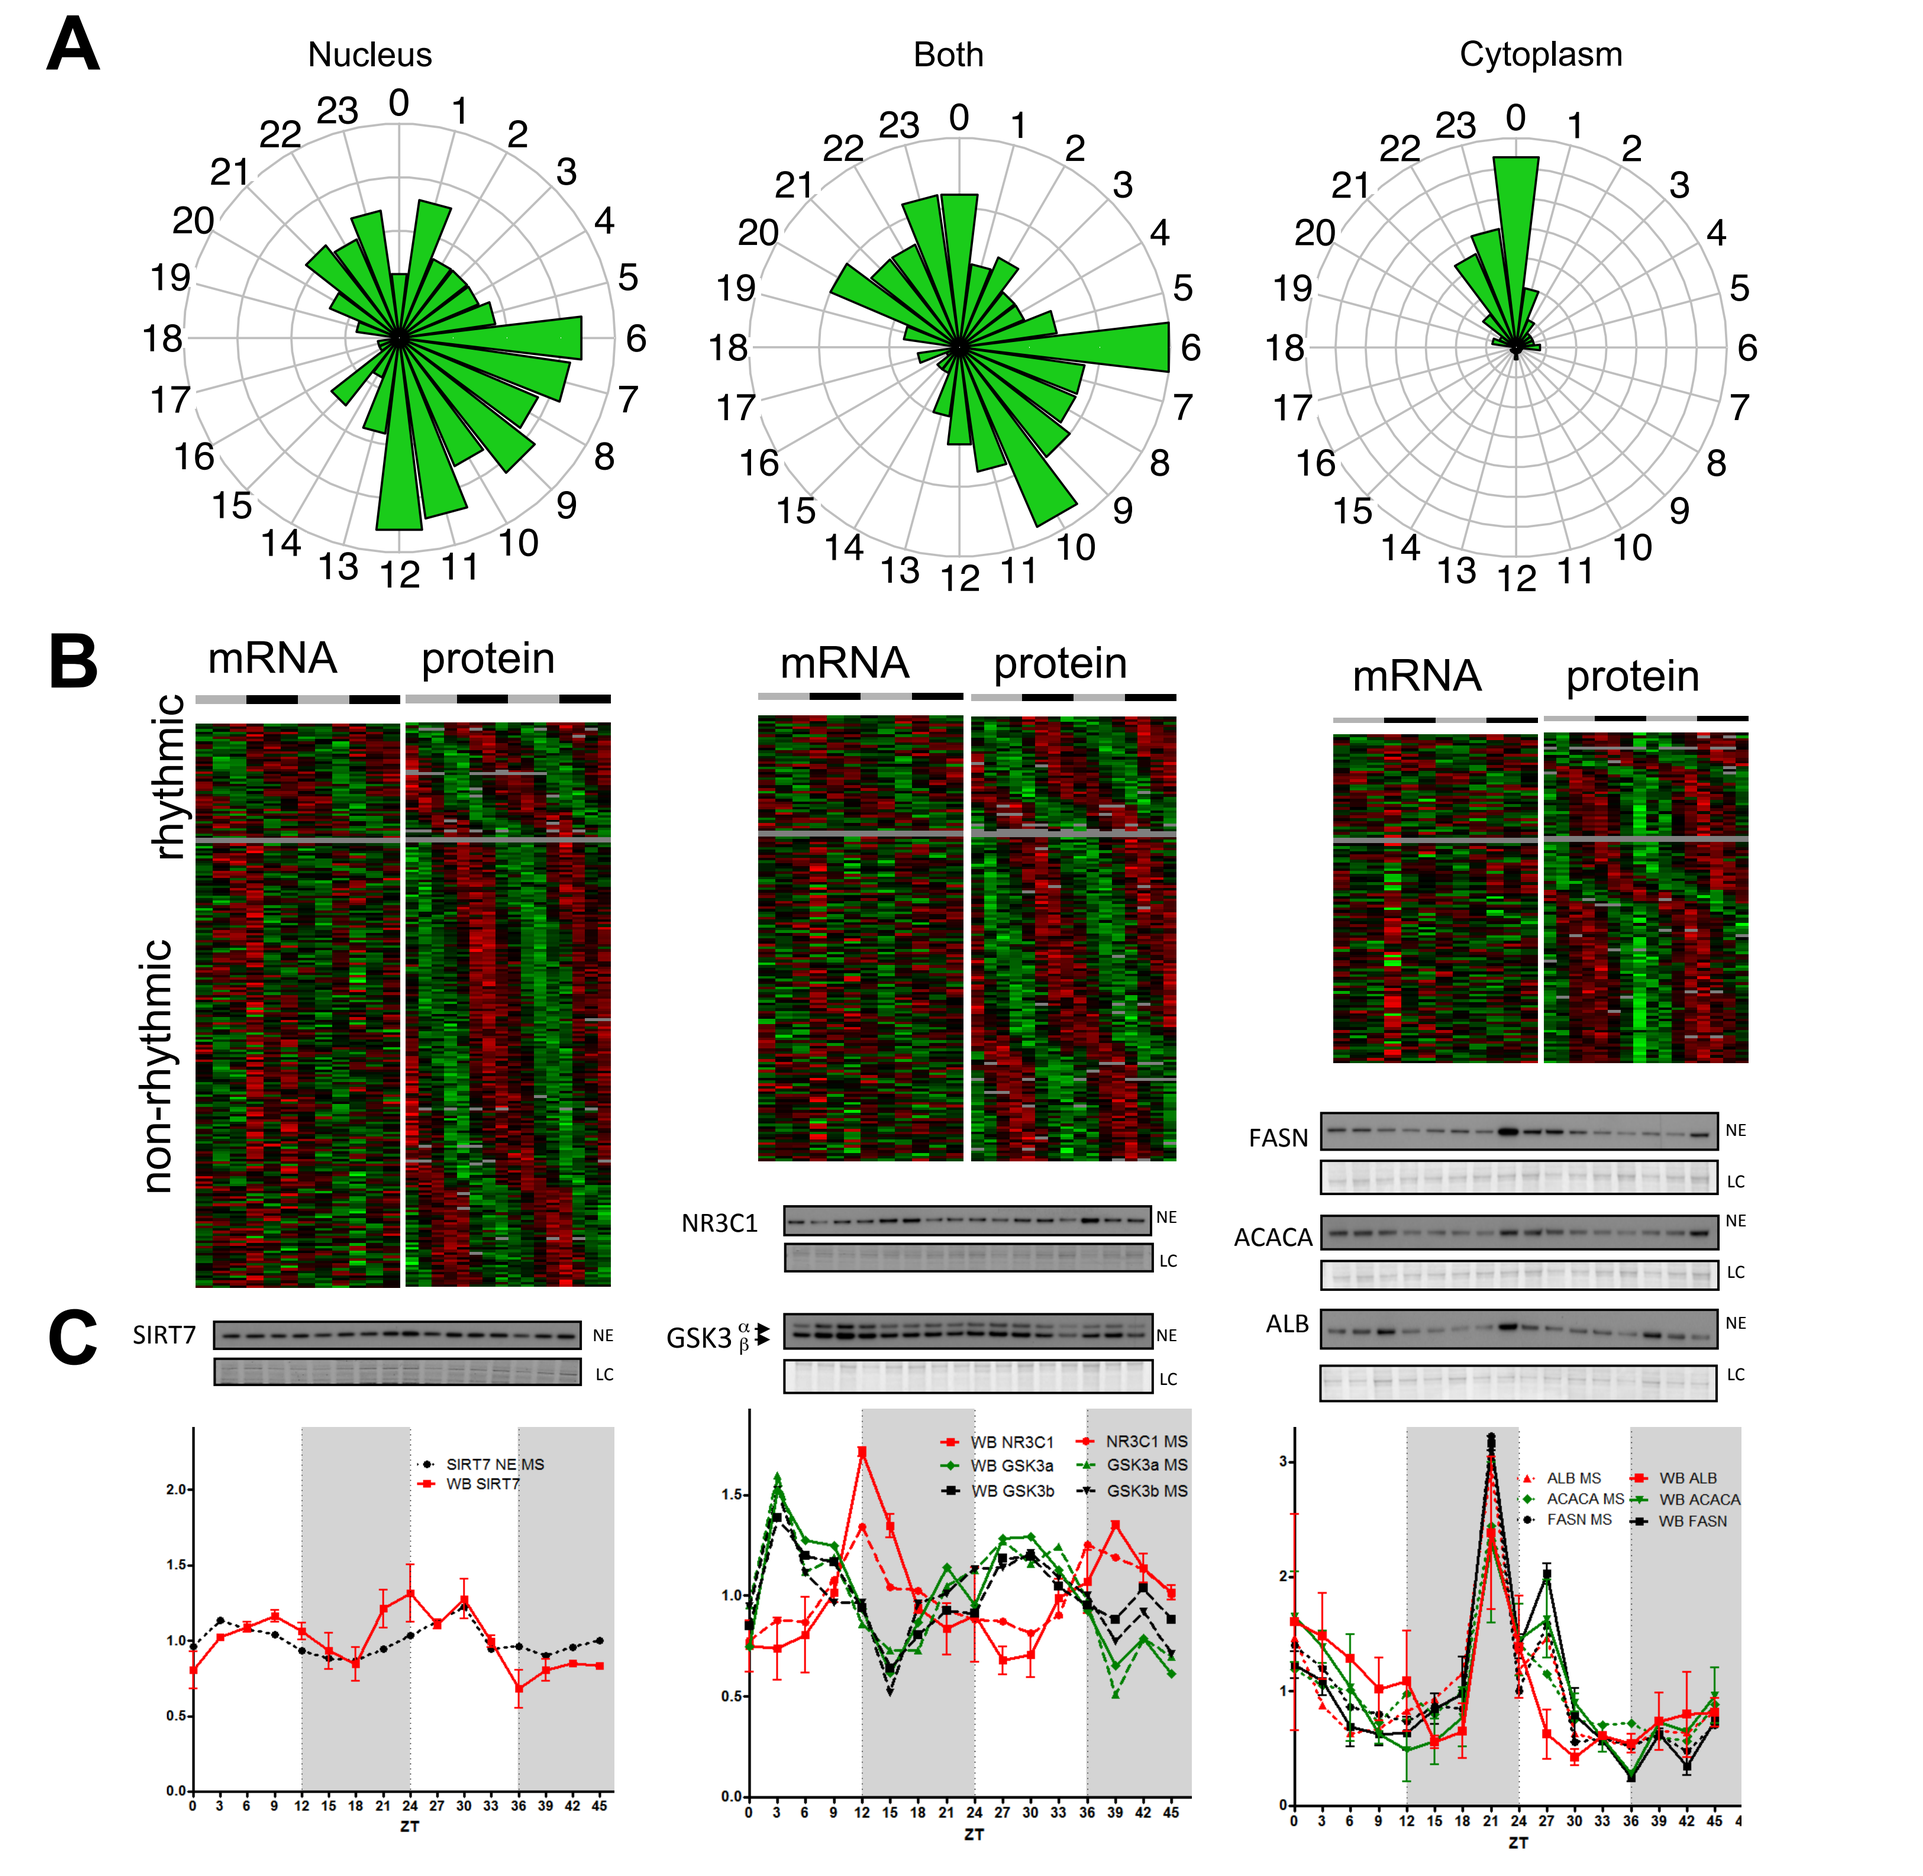
Nucleus
Both
Cytoplasm
mRNA
protein
rhythmic
non-rhythmic
SIRT7
NR3C1
GSK3
FASN
ACACA
ALB

A) Phase distribution for the rhythmic nuclear proteins (FDR<0.05) grouped by their annotated localizations (UNIPROT)
B) Heat maps of the rhythmic proteins and their corresponding mRNAs. Data is standardized by rows and gray blocks indicate missing data.
C) Western blot of individual rhythmic proteins performed on nuclear extract. The graphs represents the quantification of the blots and the corresponding mass spec data.

Fig. 3 Subunits of nuclear protein complexes showed highly similar and diurnal accumulations

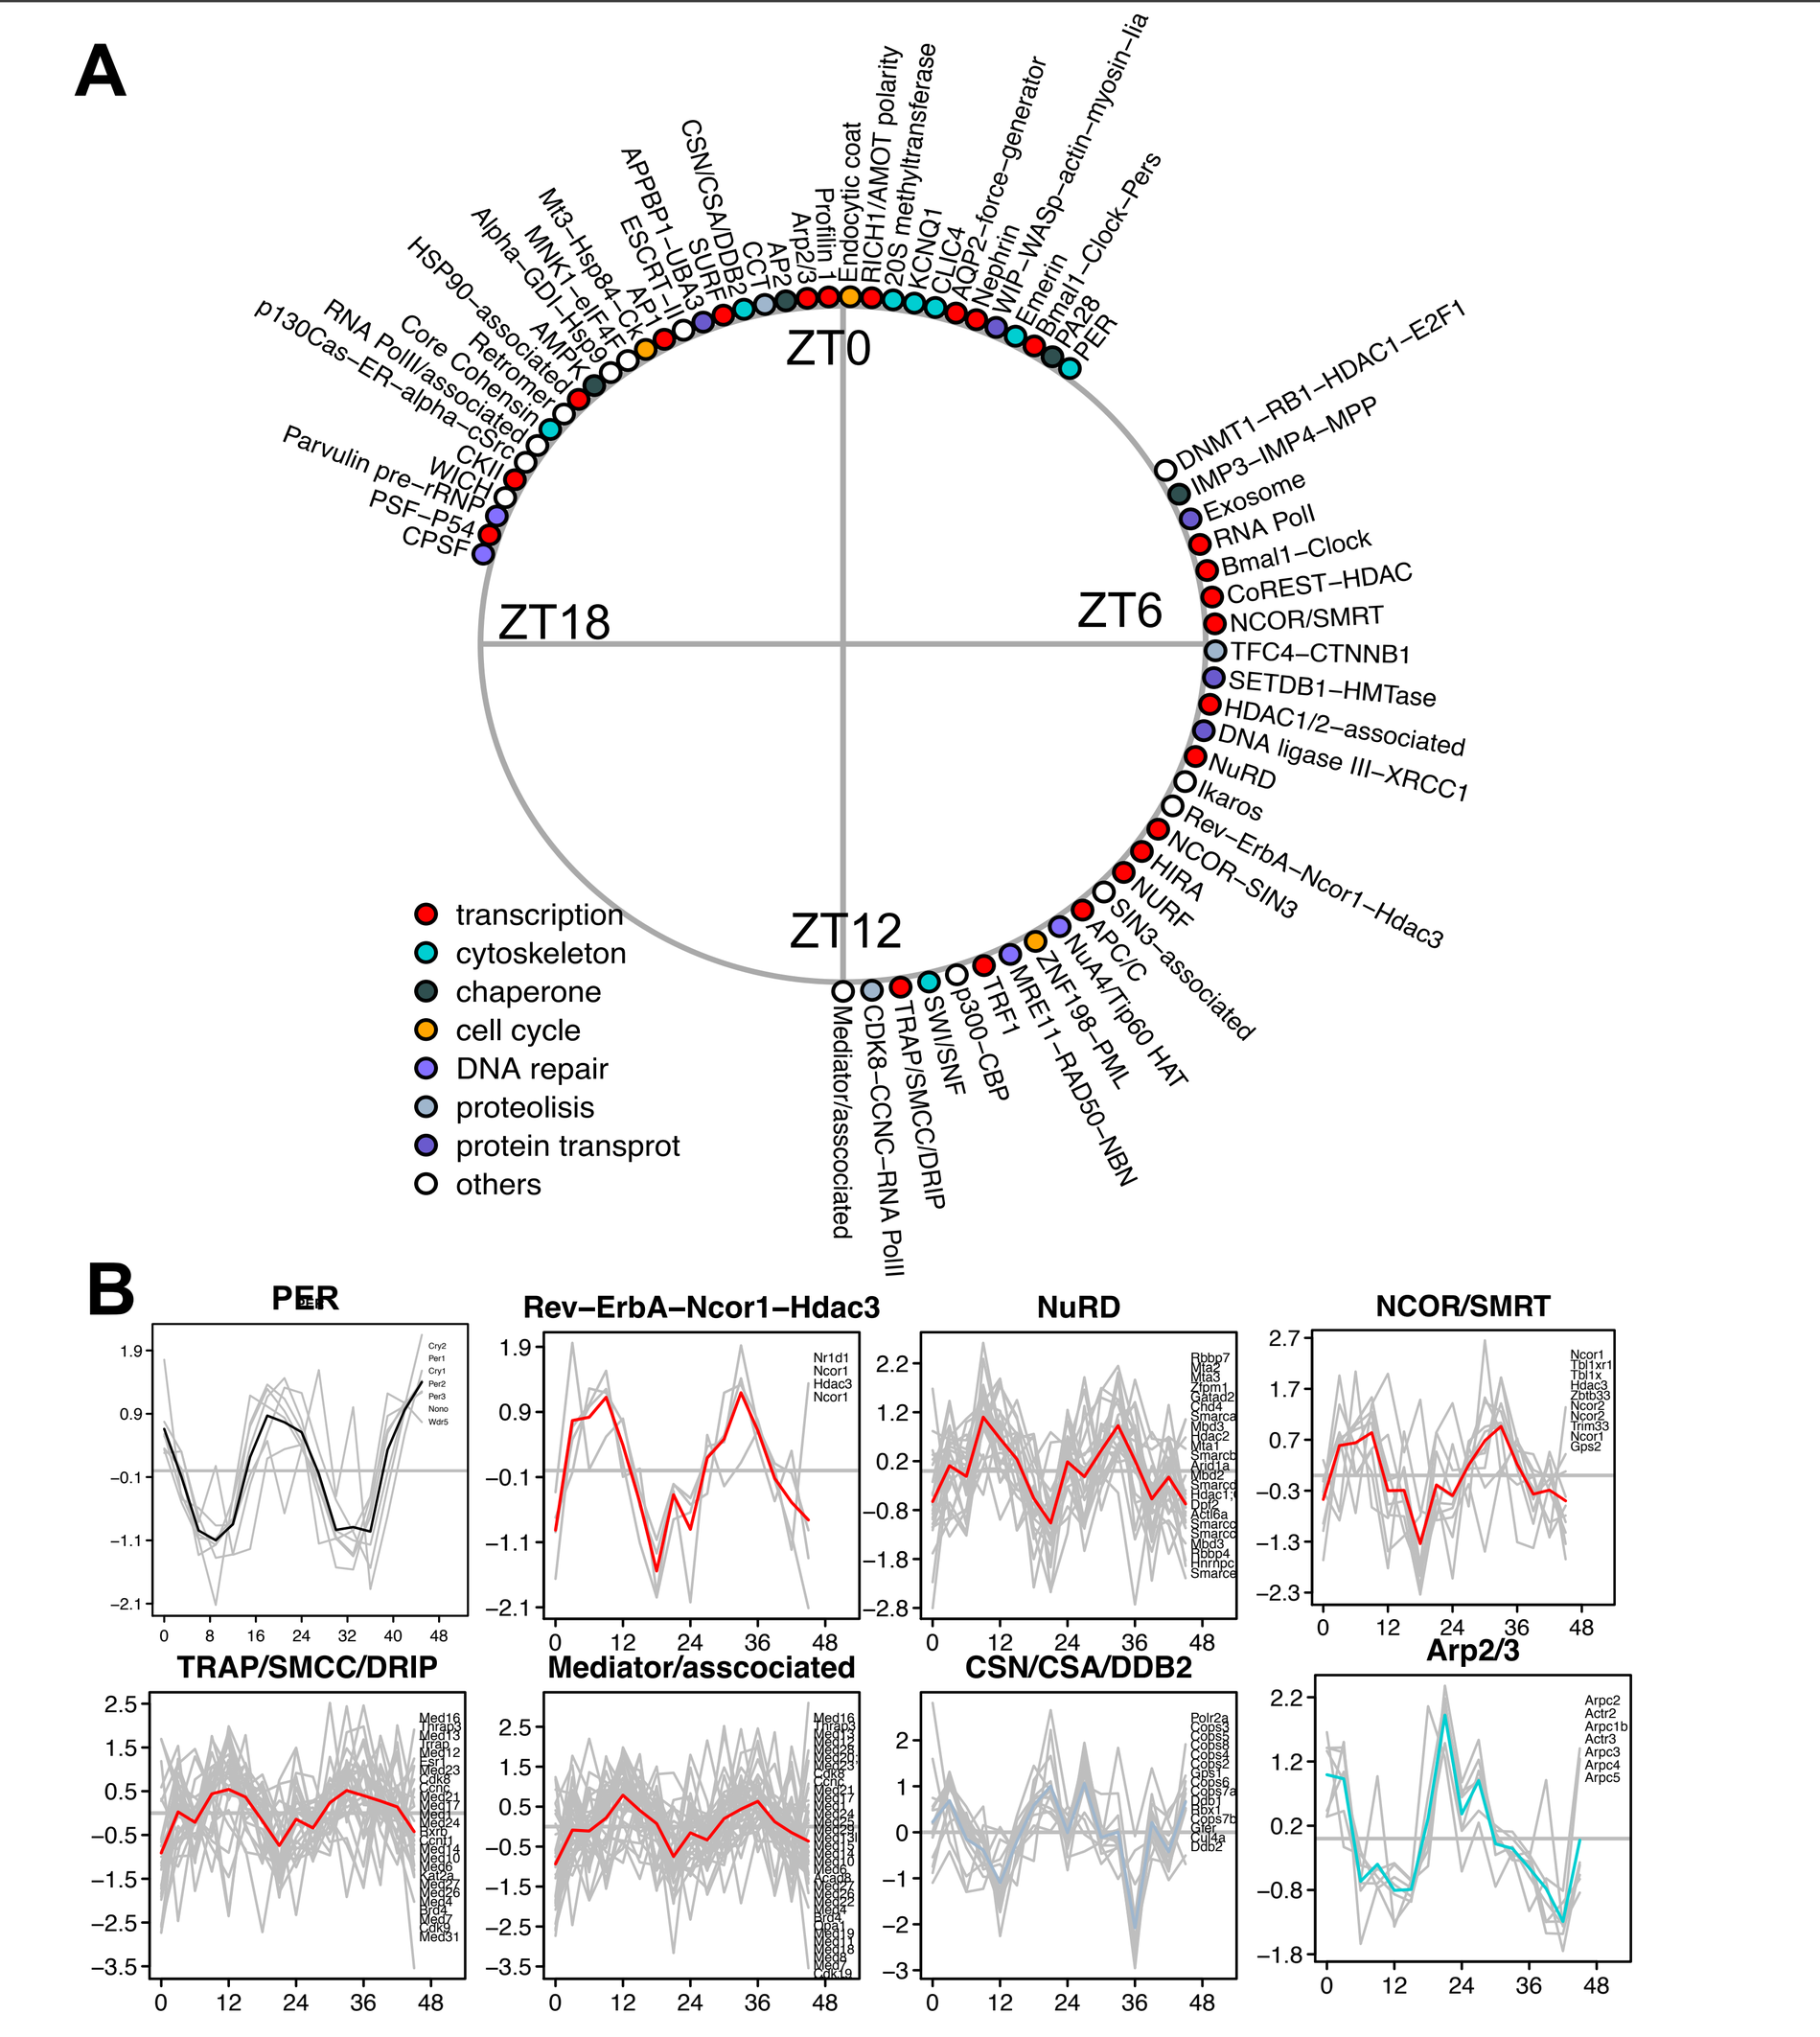
ZT0
ZT6
ZT12
ZT18
transcription
cytoskeleton
chaperone
cell cycle
DNA repair
proteolisis
protein transprot
others
PER
Rev−ErbA−Ncor1−Hdac3
NuRD
NCOR/SMRT
TRAP/SMCC/DRIP
Mediator/asscociated
CSN/CSA/DDB2
Arp2/3

A) Peak phases of rhythmic nuclear protein complexes and individual examples are found in **B).**

Fig.4 Rhythmic activities of kinases predicted by nuclear phospho-proteome

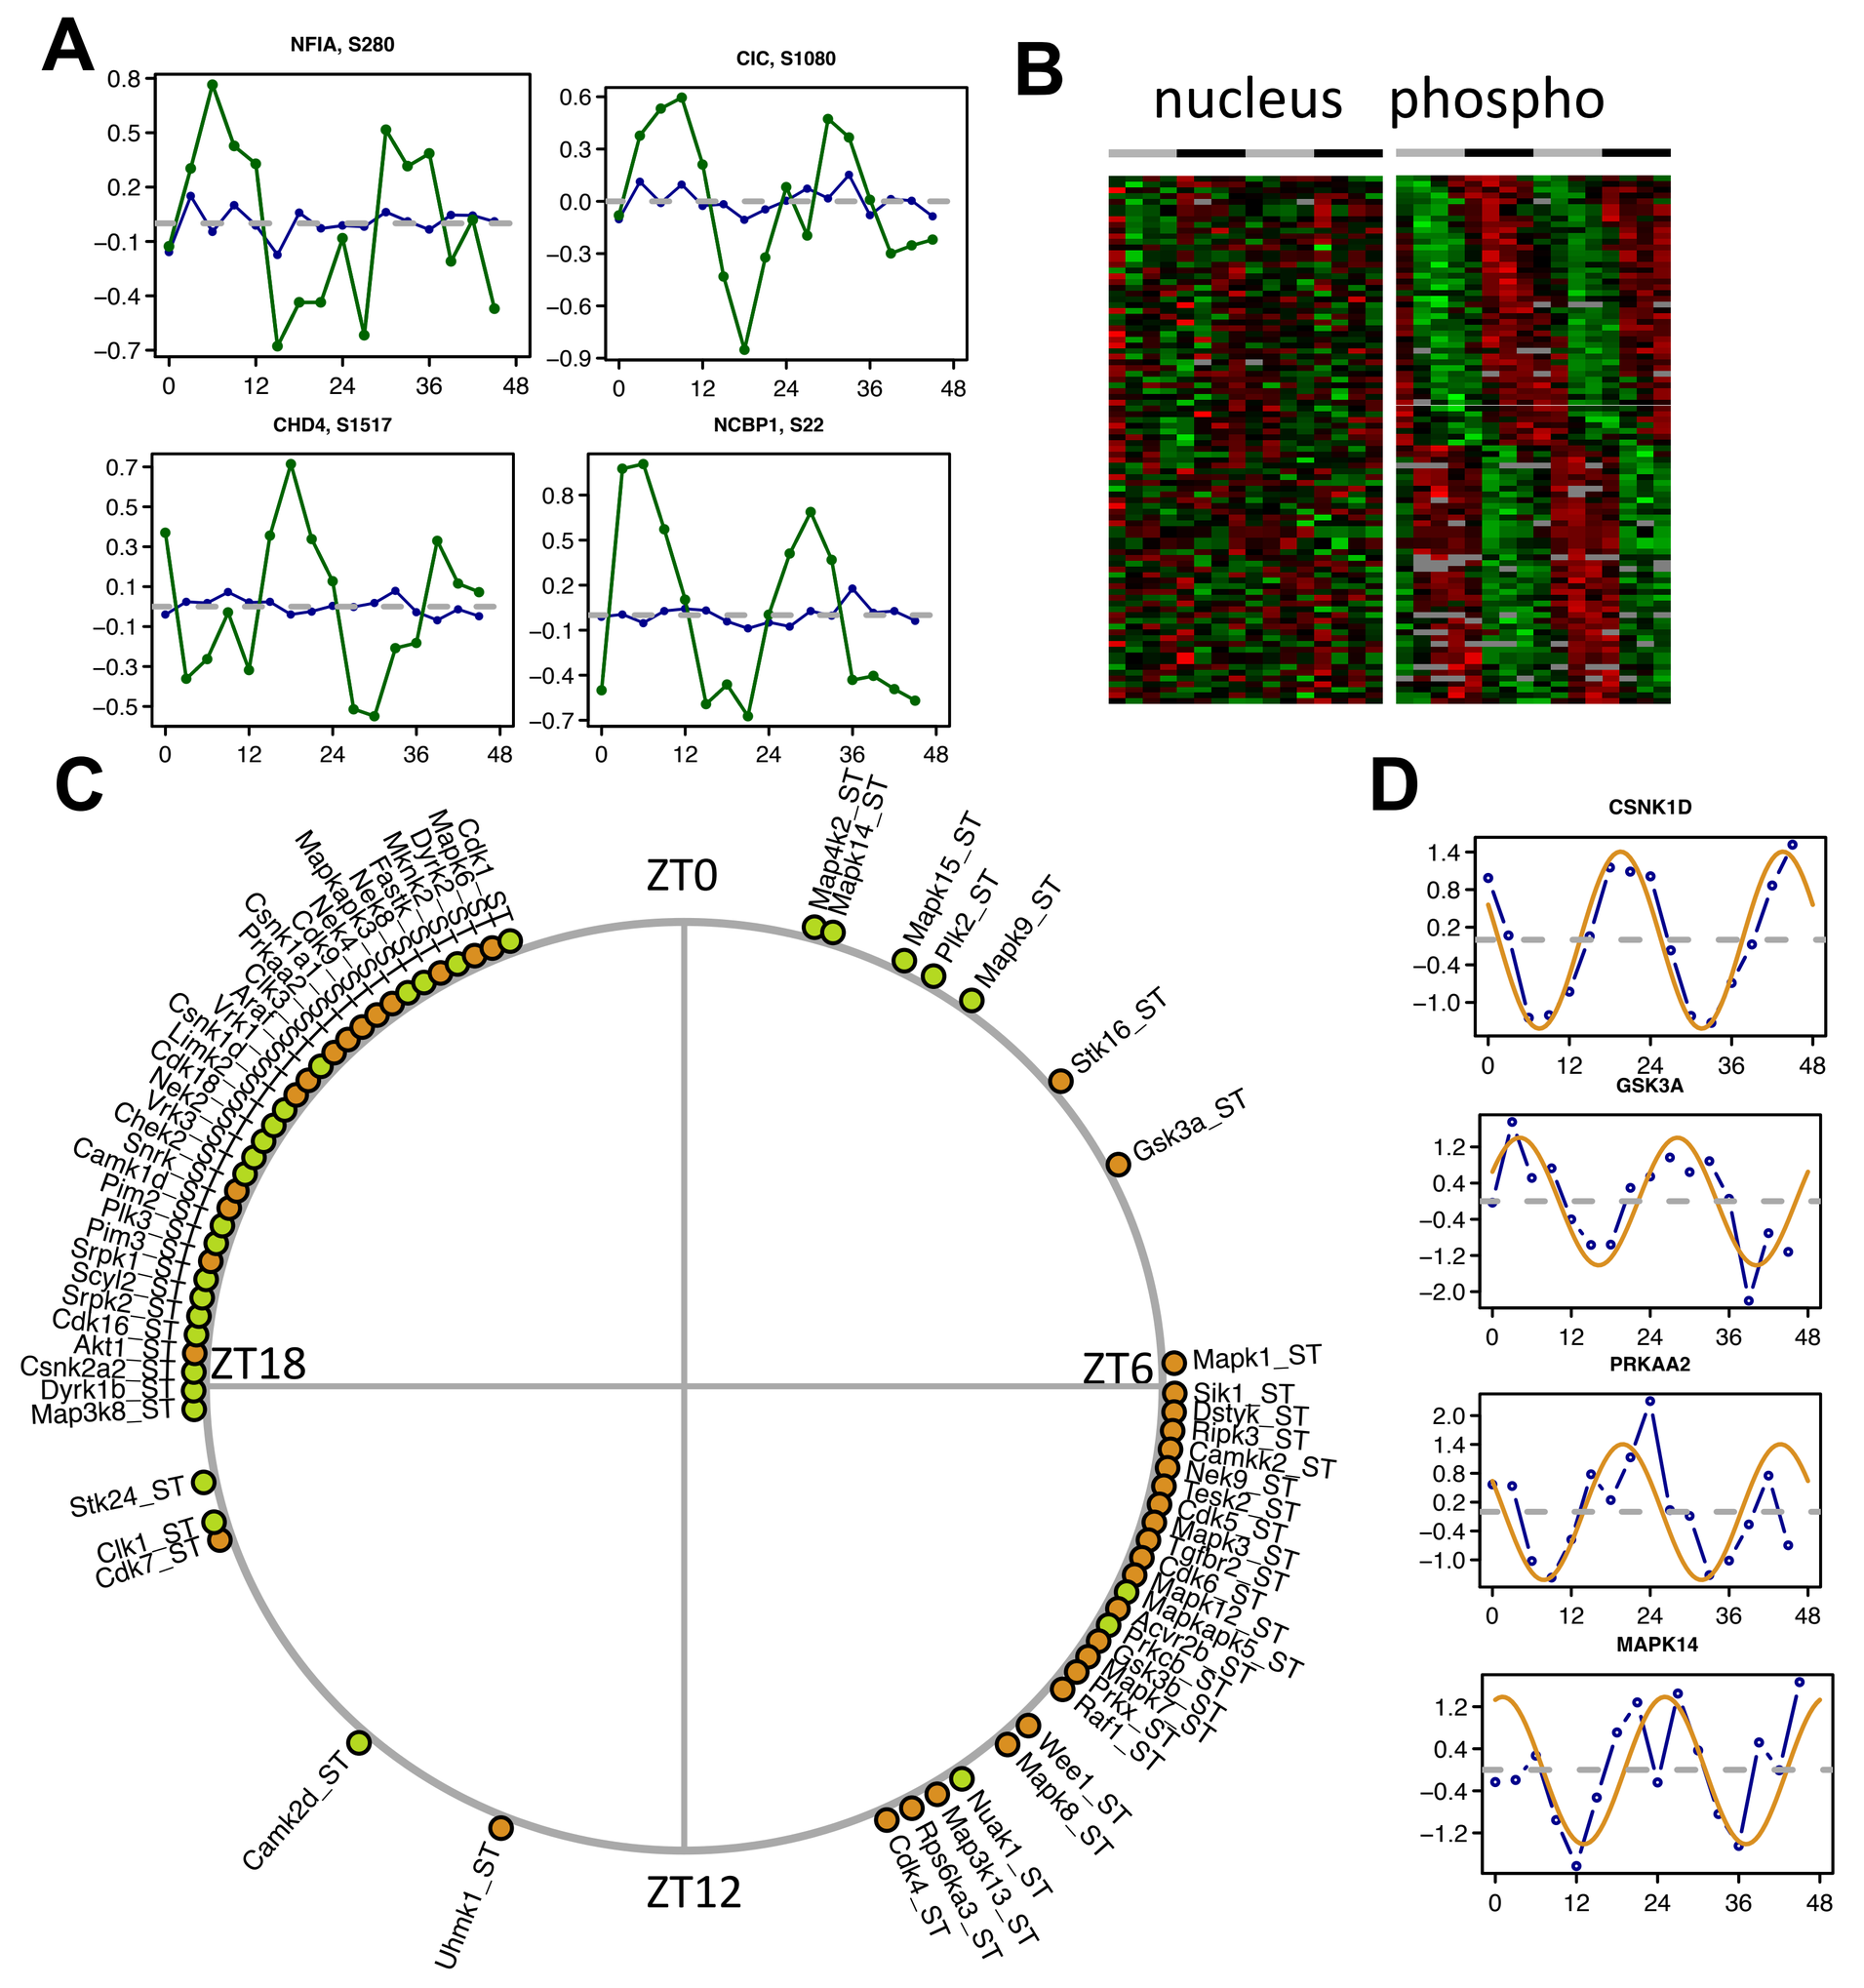
NFIA, S280
CIC, S1080
CHD4, S1517
NCBP1, S22
nucleus
phospho
ZT0
ZT6
ZT12
ZT18
CSNK1D
GSK3A
PRKAA2
MAPK14

A) and **B)** Examples and heat maps of rhythmic nuclear phospho-proteins with non-rhythmic nuclear proteins.
C) Peak phases of rhythmic kinase activities predicted by the nuclear phospho-proteins, some of which are confirmed by their nuclear protein accumulations in **D)**

Fig. 5 Transcription factors and coregulators (FDR<5%) involved in the circadian transcription

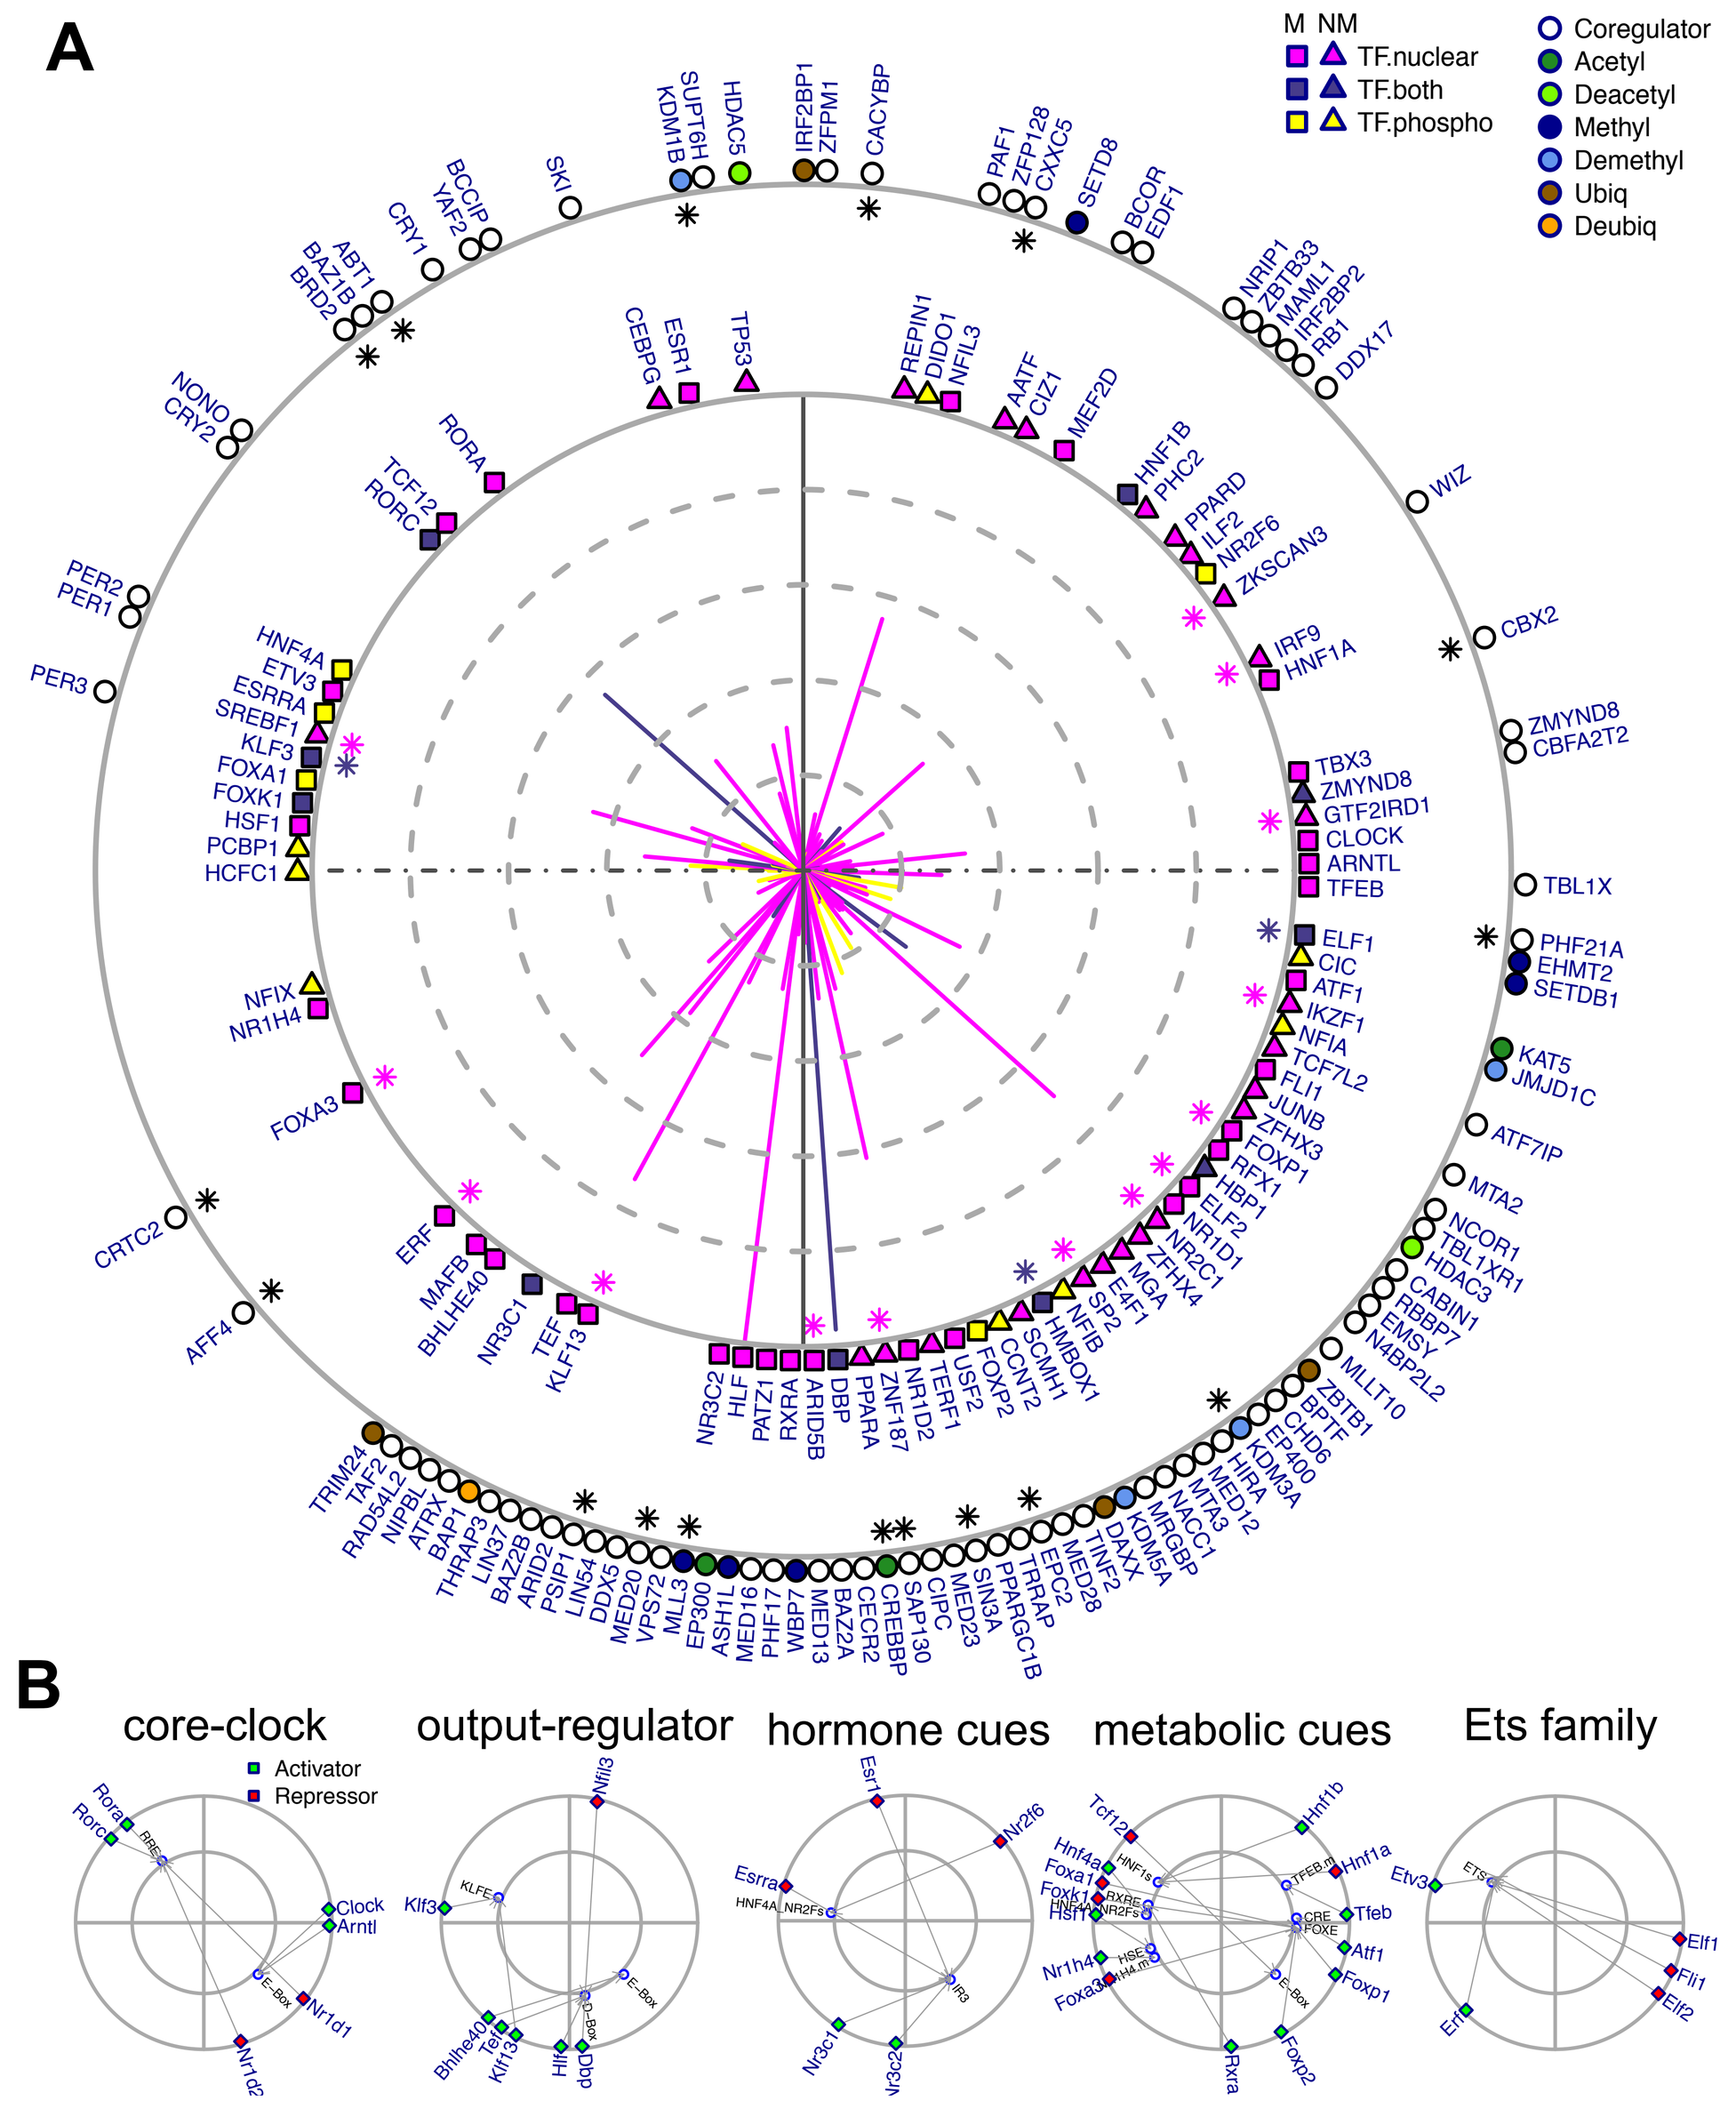
TF.nuclear
TF.both
TF.phospho
Coregulator
Acetyl
Deacetyl
Methyl
Demethyl
Ubiq
Deubiq
core-clock
Activator
Repressor
output-regulator
hormone cues
metabolic cues
Ets family

Peak phases of TF accumulations vs. those of predicted motifs

Fig. 6 Rhythmic endoreplication probably causes leakage of cytoplasmic proteins by weakening nuclear envelope

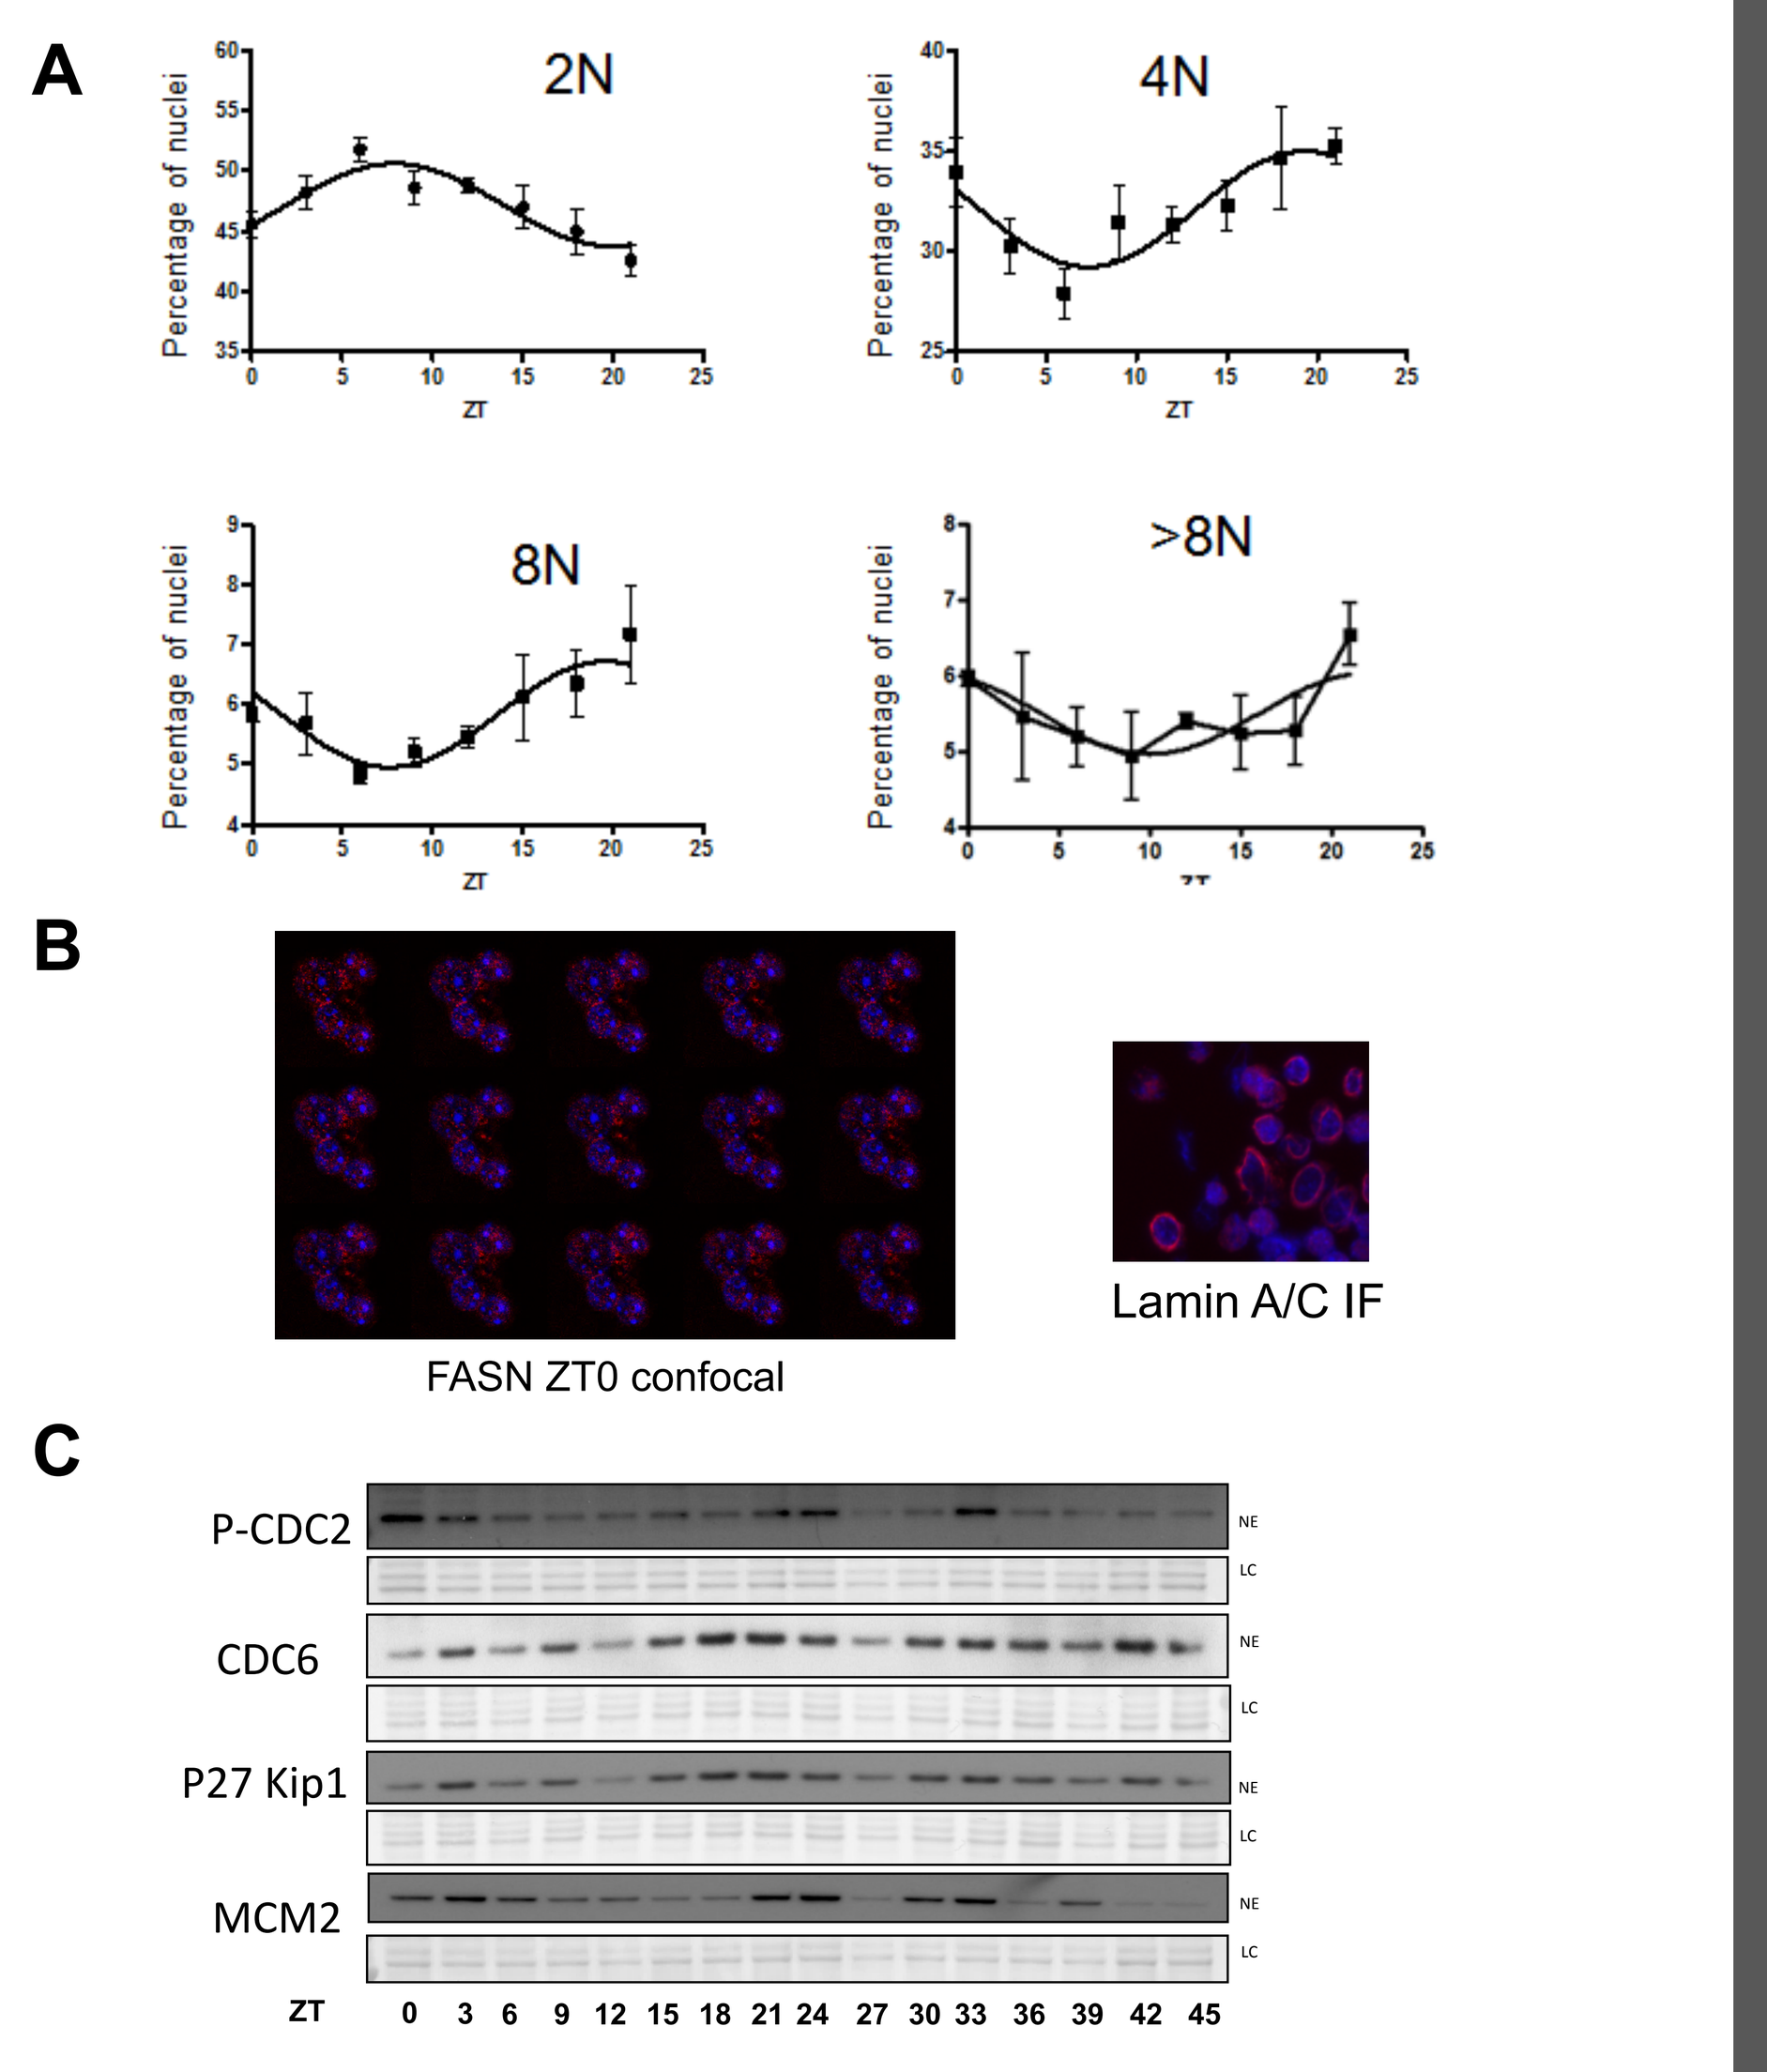
2N
4N
8N
>8N
Percentage of nuclei
ZT
FASN ZT0 confocal
Lamin A/C IF
P-CDC2
CDC6
P27 Kip1
MCM2
ZT
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45

A) FACS analyses of nuclei isolated from mouse liver around the clock and separated into ploidy populations by (n = 4 light dark cycles)
B) Localization of FASN at ZT0 (confocal fluorescence) and immunofluorescence of Lamin A/C in purified liver nuclei.
C) Western blot of individual protein involved in cell cycle regulation and DNA replication.

Take-home messages

1. SILAC nuclear proteomics in mouse liver
 - High resolution (~5000 proteins),
 - ~12% highly rhythmic (FDR<5%).
 - Almost all core clock and clock related genes identified and quantified.
2. Within these rhythmic proteins
 - 81 transcription factors and more than 100 coregulators : most of them are new.
 - Many nuclear protein complexes also display diurnal expression.

3.Annotated cytoplasmic proteins are also

- detected in the nucleus with a sharp day night transition phase.

➔ may be due to a weakening of the nuclear envelope resulting from rhythmic endoreplication/replication.